MYRIAD RBM®

Analysis of Protein Biomarkers in Prostate and Colorectal Tumor Lysates

Laurie L. Stephen, Karri L. Ballard, Marc Damour, Josh Kemp, Ralph L. McDade, James P. Mapes.

Introduction

Traditional tumor biomarkers are proteins that have been shed in the bloodstream and have been critical in identifying and treating cancer. Myriad RBM and the Cancer Prevention Research Institute of Texas (CPRIT) have continued their collaborative efforts to develop new assays for cancer biomarkers on the Myriad RBM Multi-Analyte Profile (MAP) service platform. The release of OncologyMAP® 2.0 expanded Myriad RBM's biomarker menu to over 250 analytes, which can be measured from a small single sample. Using this comprehensive menu, we surveyed proteins relevant to oncology in colon and prostate tumors in order to identify potential proteins of interest for cancer research.

Aims and Methods

- In Exp 1, 4 colorectal (CRC) and 3 prostate (PC) tumors were purchased from Proteogenex (Culver City, CA). Samples were collected and flash frozen in liquid nitrogen and stored at -80°C.
- Two sections of each tumor were homogenized in RIPA buffer with protein inhibitors at Myriad RBM and analyzed using OncologyMAP 2.0 to measure 125 analytes.
- All immunoassays were run using the Luminex Technology with Tecan automation. Assay validation was guided by CLSI recommendations
- In Experiment 2, CRC tumor, adjacent tissue and serum from the same individual (n=6) were purchased and processed as described above.

Results and Discussion

Fig. 1 Analyte Changes (log/log) in Colorectal Cancer Compared to Prostate Cancer

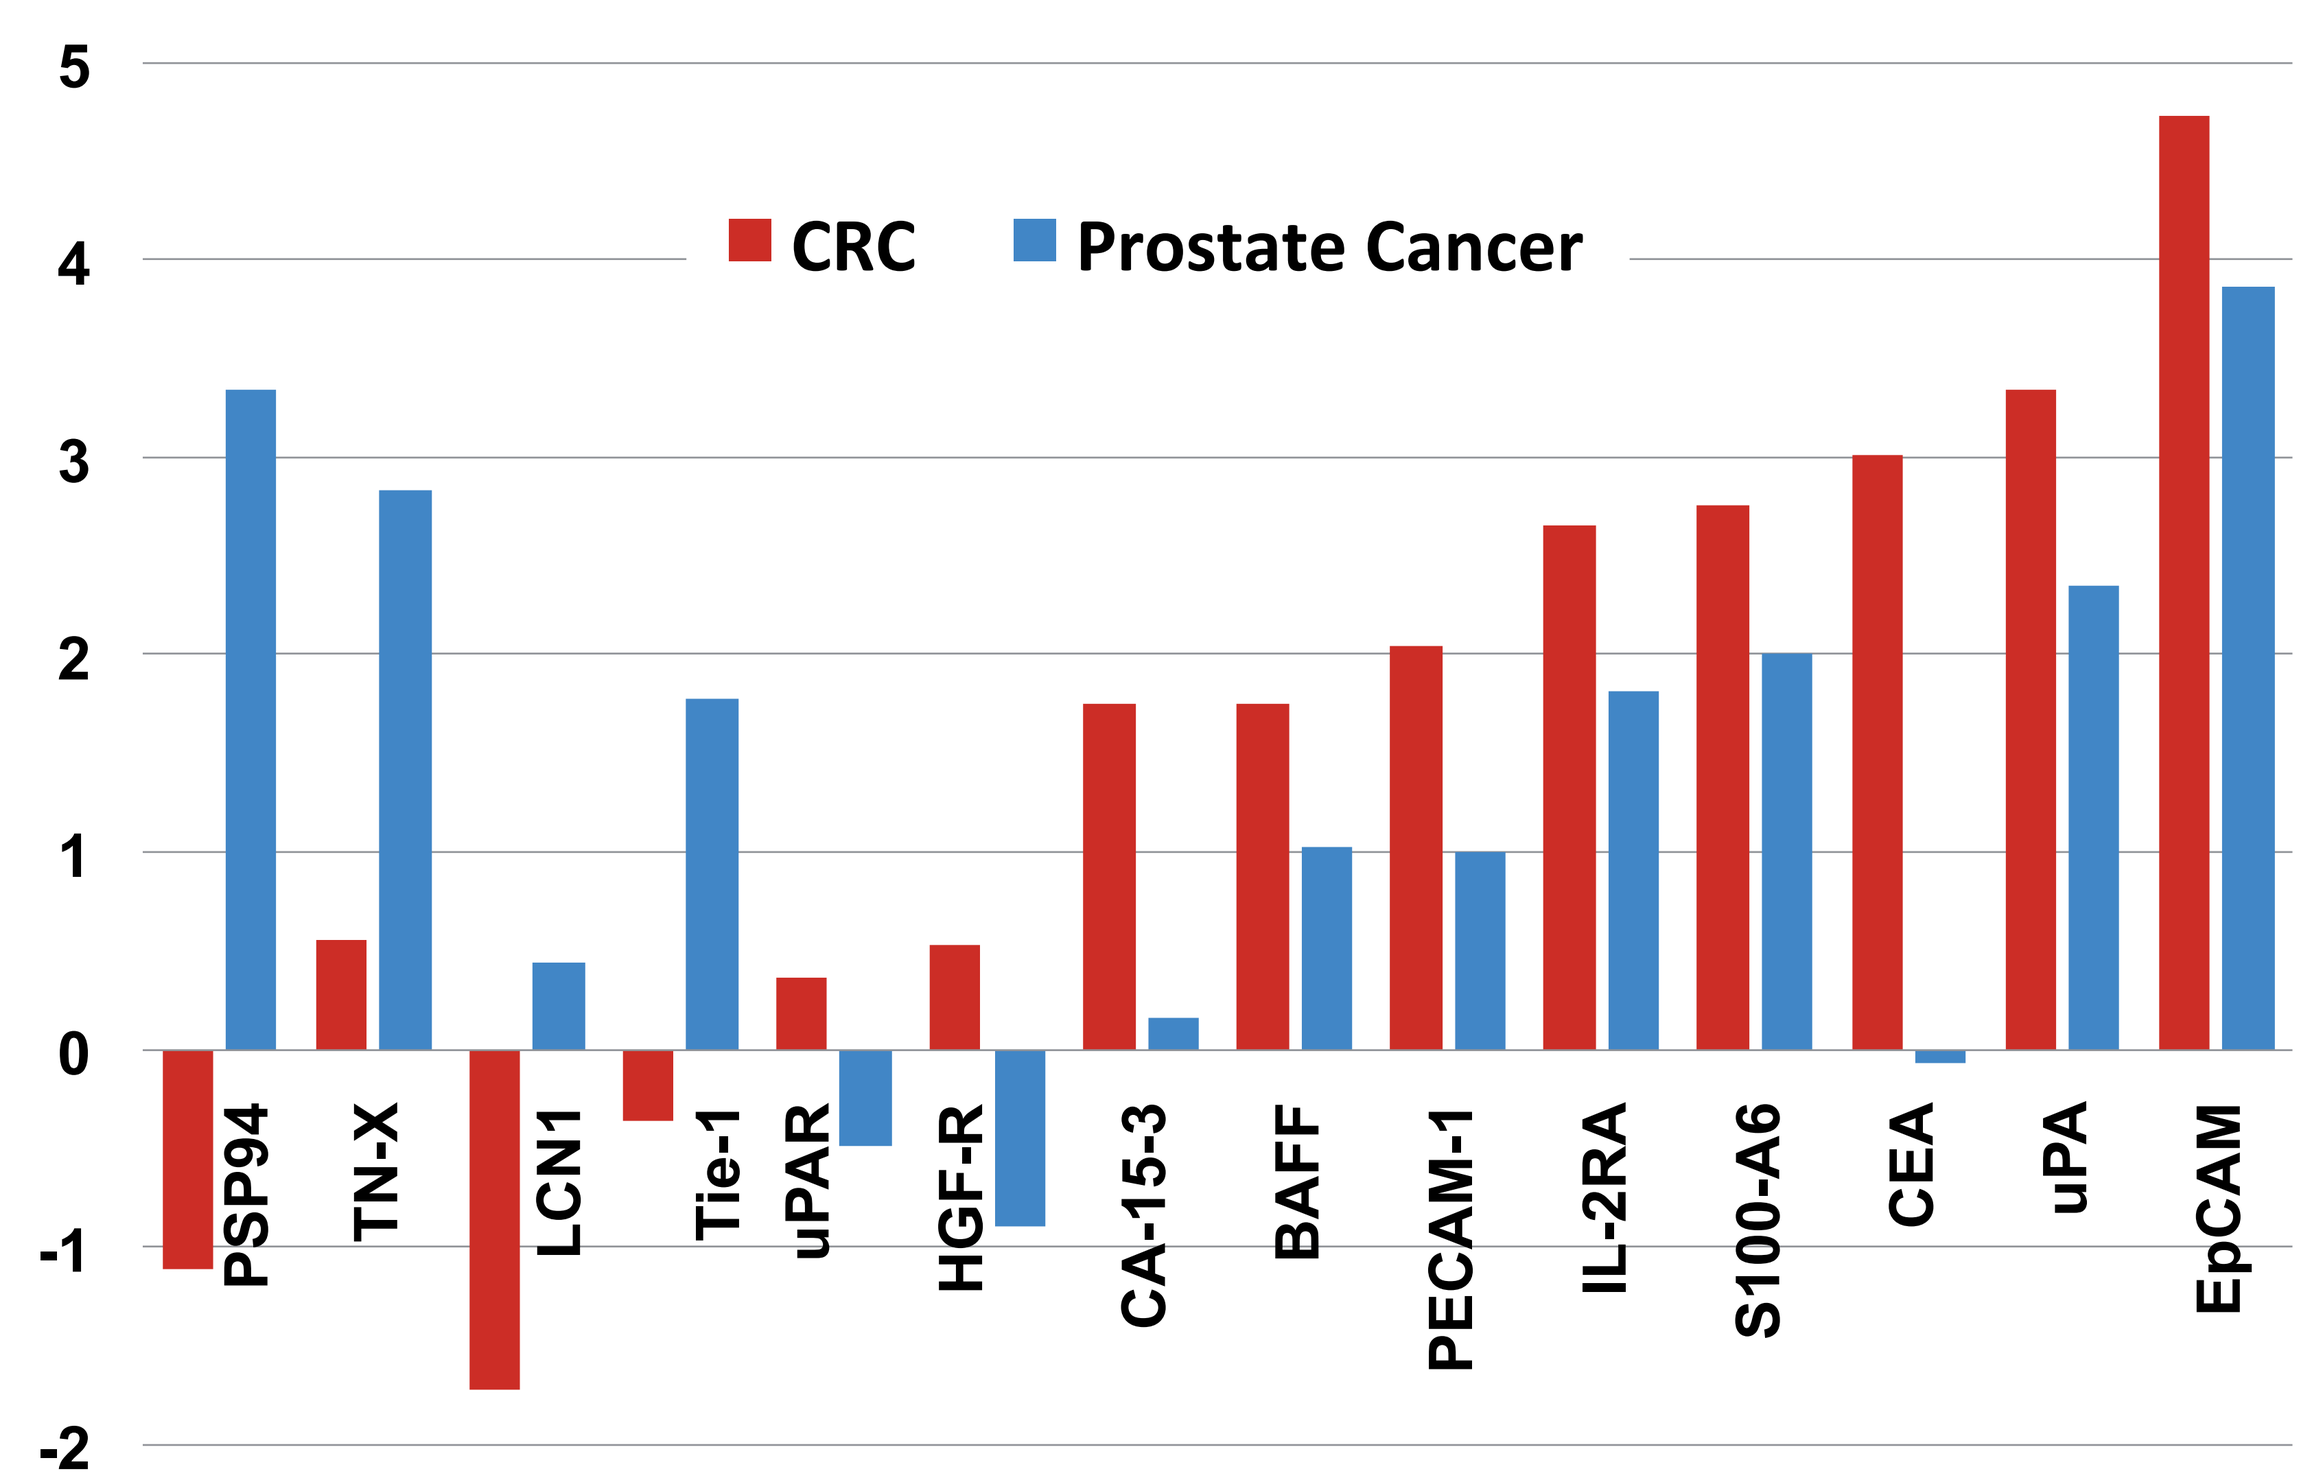
CRC
Prostate Cancer
5
4
3
2
1
0
-1
-2
PSP94
TN-X
LCN1
Tie-1
uPAR
HGF-R
CA-15-3
BAFF
PECAM-1
IL-2RA
S100-A6
CEA
uPA
EpCAM

Table 1. Fold change of concentration of prostate tumor vs. CRC tumor homogenate

Analyte	Fold Change	P Value
PSP94	(28,368)	0.0017
TN-X	(187)	0.0003
LCN1	(147)	0.0003
Tie-1	(135)	0.0069
uPAR	7	0.0052
HGF-R	27	0.0056
CA-15-3	38	0.0071
BAFF	5	0.0081
PECAM-1	11	0.0054
IL-2RA	7	0.0000
S100-A6	6	0.0047
CEA	1,224	0.0043
uPA	10	0.0012
EpCAM	7	0.0044

Fig 2. Homogeneity of CRC tumor measurements

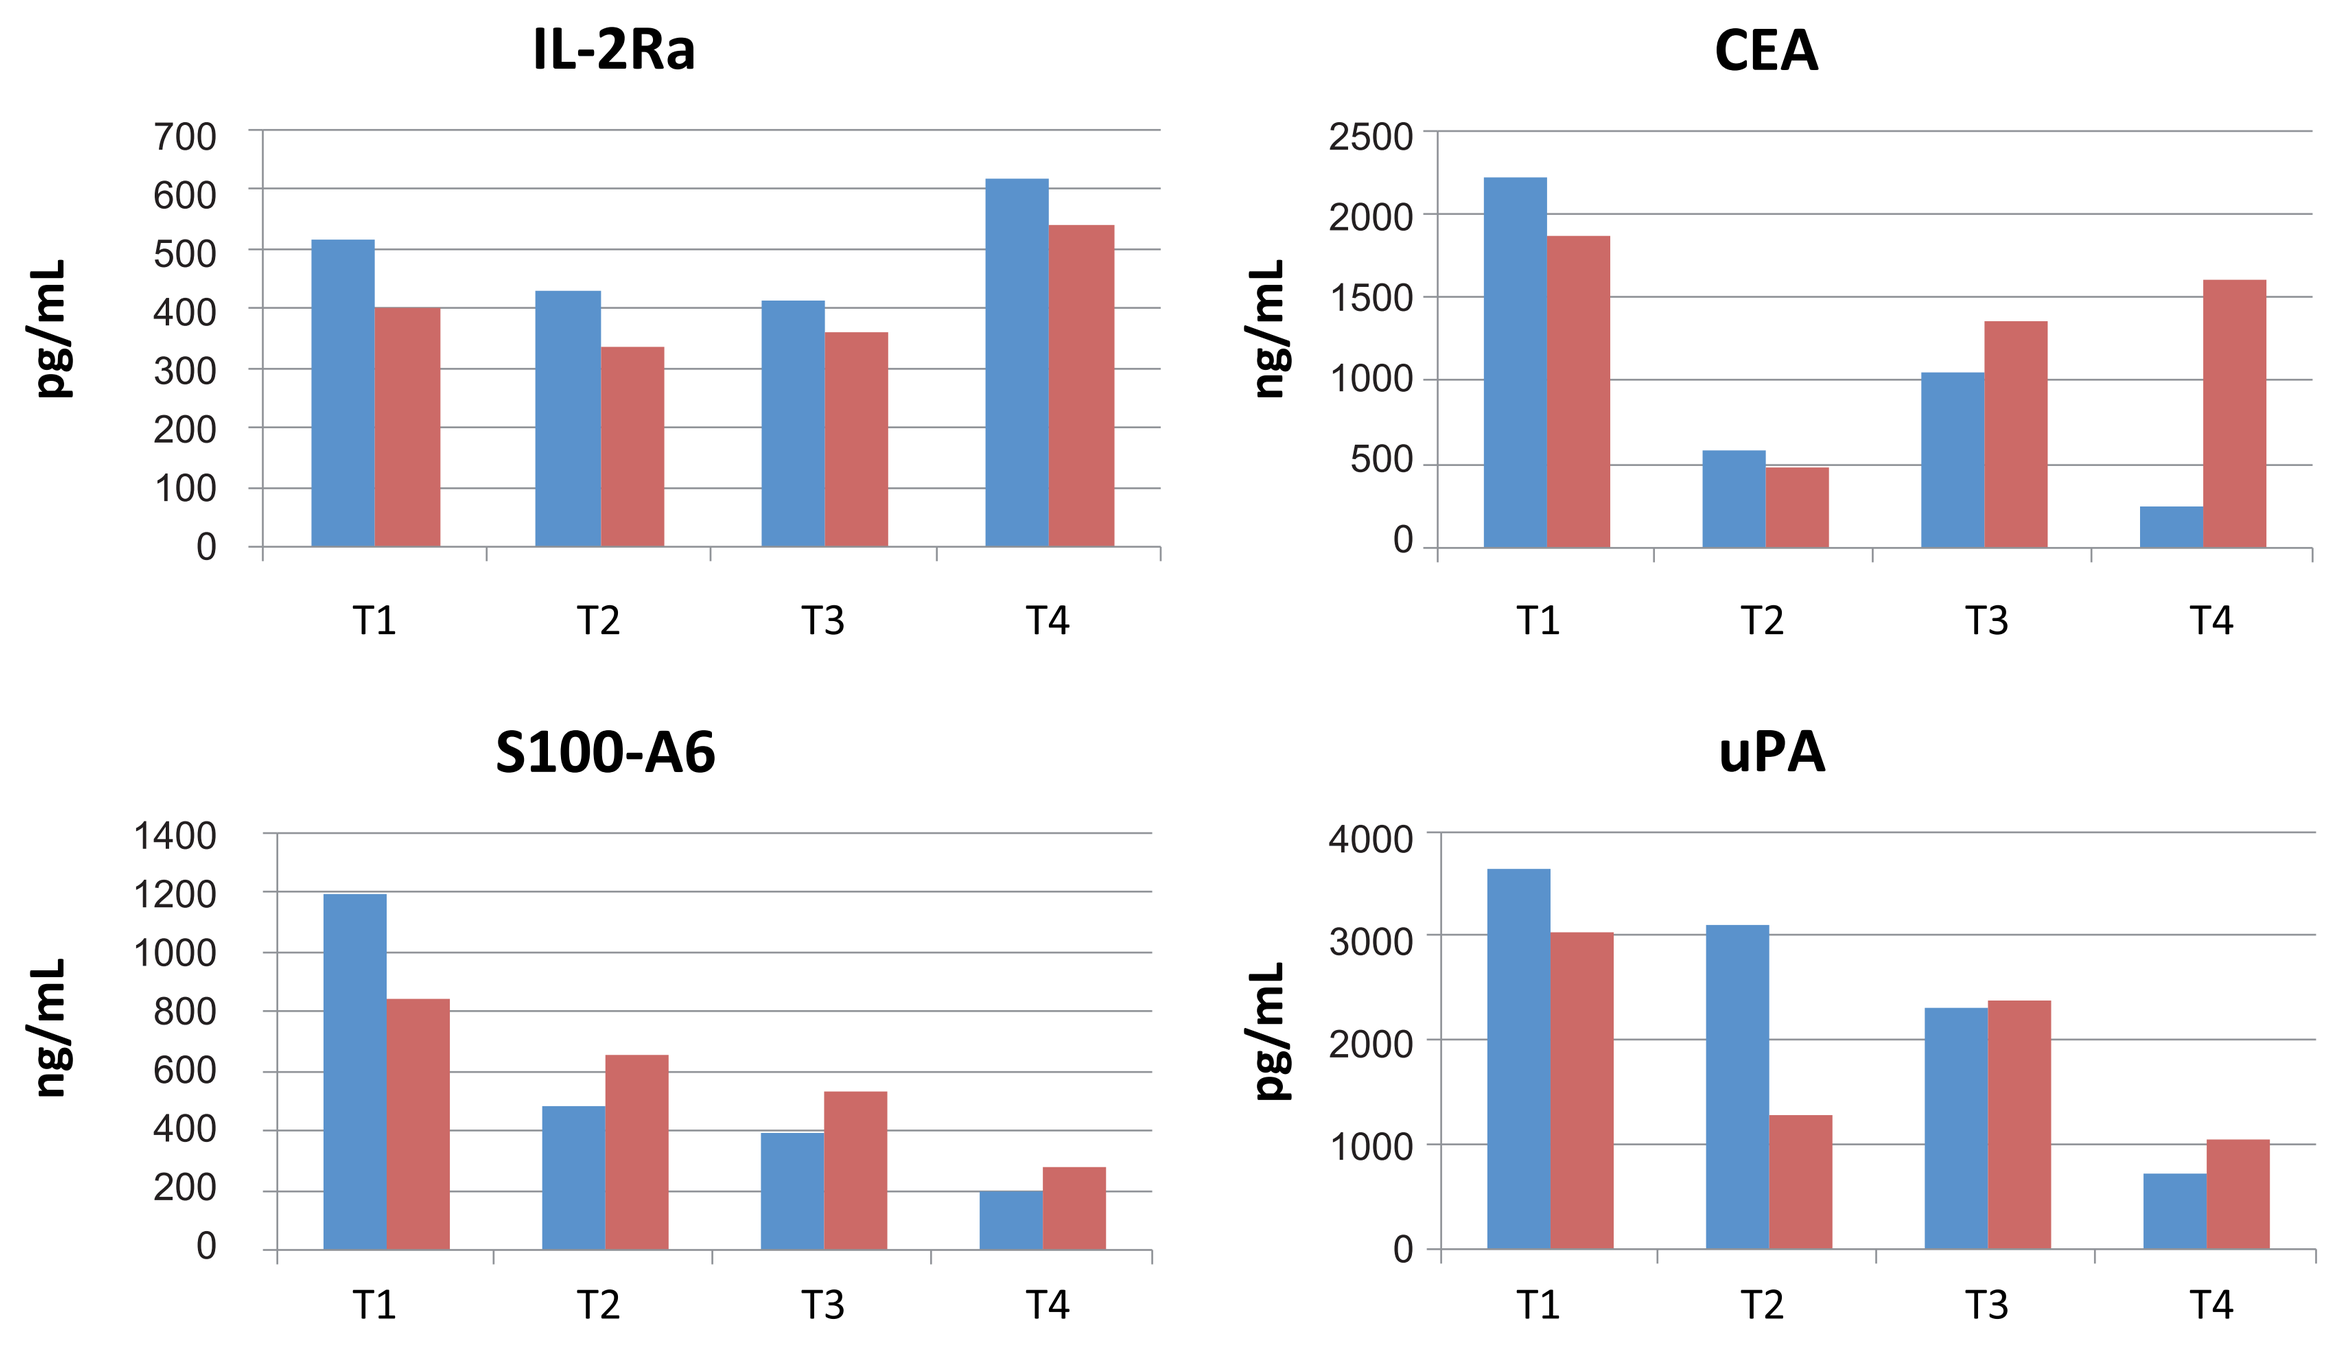
IL-2Ra
pg/mL
700
600
500
400
300
200
100
0
T1
T2
T3
T4
CEA
ng/mL
2500
2000
1500
1000
500
0
T1
T2
T3
T4
S100-A6
ng/mL
1400
1200
1000
800
600
400
200
0
T1
T2
T3
T4
uPA
pg/mL
4000
3000
2000
1000
0
T1
T2
T3
T4

Table 2. Statistically Significant Differences in CRC versus Adjacent Tissue

		Adjacent Tissue	Colorectal Cancer Tissue	P Value
Analytes	Units	Mean		t-test
Monocyte Chemotactic Protein 1 (MCP-1)	pg/mL	13	106	2.8E-05
Urokinase-type Plasminogen Activator (uPA)	pg/mL	278	1660	1.9E-04
Carcinoembryonic Antigen (CEA)	ng/mL	96	2444	2.3E-04
Tissue type Plasminogen activator (tPA)	ng/mL	2.0	0.89	4.0E-04
Fibroblast Growth Factor basic (FGF-basic)	pg/mL	610	171	5.5E-04
YKL-40	ng/mL	0.40	2.6	7.0E-04
Vascular Endothelial Growth Factor (VEGF)	pg/mL	49	312	9.9E-04
Tenascin-X (TN-X)	ng/mL	6.1	2.4	1.7E-03
Cathepsin B (pro) (CTSB)	ng/mL	1.7	4.2	1.7E-03
Interferon gamma Induced Protein 10 (IP-10)	pg/mL	60	599	2.1E-03
Collagen IV	ng/mL	361	1919	2.8E-03
Neuron-Specific Enolase (NSE)	ng/mL	18	4.7	2.9E-03
Interleukin-6 receptor subunit beta (IL-6R beta)	ng/mL	1.5	0.76	4.9E-03
Carcinoembryonic antigen-related cell adhesion molecule 6 (CEACAM6)	ng/mL	195	8421	5.1E-03
Human Chorionic Gonadotropin beta (hCG)	mIU/mL	0.84	1.4	5.5E-03
Decorin	ng/mL	138	18	5.6E-03
Endoglin	ng/mL	0.031	0.075	6.4E-03
Carbonic anhydrase 9 (CA-9)	ng/mL	1.5	15	6.7E-03
Cadherin-13 (T-cad)	ng/mL	3.2	2.0	6.9E-03
Fatty Acid-Binding Protein, adipocyte (FABP, adipocyte)	ng/mL	77	15	8.3E-03
6Ckine	pg/mL	67	179	1.1E-02
Neutrophil Gelatinase-Associated Lipocalin (NGAL)	ng/mL	28	117	1.3E-02
Vascular Endothelial Growth Factor Receptor 1 (VEGFR-1)	pg/mL	1517	3487	1.5E-02
Lumican	ug/mL	0.27	0.11	2.0E-02
Interferon-inducible T-cell alpha chemoattractant (ITAC)	pg/mL	41	274	3.0E-02
Complement component C1q receptor (C1qR1)	ug/mL	0.085	0.13	3.2E-02
Cellular Fibronectin (cFib)	ug/mL	0.58	3.0	3.4E-02
Lactoferrin (LTF)	ng/mL	12	26	4.0E-02
Cystatin-A	ng/mL	1.5	3.0	4.7E-02
Betacellulin (BTC)	pg/mL	179	98	4.8E-02

Table 3. Correlation of [analyte] in Serum to CRC Tumor

Analytes				Serum to Tumor
	Units	Serum	Tumor	Correlation r^2
Hepsin	pg/mL	1012	28	1.0
Insulin-like Growth Factor Binding Protein 6 (IGFBP6)	ng/mL	365	1.1	1.0
Osteopontin	ng/mL	26	1.6	1.0
Lipocalin-1 (LCN1)	ng/mL	3.2	0.054	0.99
Beta-microseminoprotein (PSP94)	ng/mL	8.0	1.5	0.98
Tie-2	ng/mL	10	0.066	0.83
Maspin	pg/mL	2923	6770	0.76
Endoglin	ng/mL	2.4	0.075	0.74
Interferon gamma Induced Protein 10 (IP-10)	pg/mL	647	599	0.71
Human Chorionic Gonadotropin beta (hCG)	mIU/mL	3.0	1.4	0.71
Monocyte Chemotactic Protein 1 (MCP-1)	pg/mL	501	106	0.68
Macrophage inflammatory protein 3 beta (MIP-3 beta)	pg/mL	321	15	0.68
Lactoferrin (LTF)	ng/mL	57	26	0.64
YKL-40	ng/mL	42	2.6	0.60
Cathepsin B (pro) (CTSB)	ng/mL	55	4.2	0.58
Osteoprotegerin (OPG)	pM	6.4	18	0.56
Urokinase-type Plasminogen Activator (uPA)	pg/mL	905	1660	0.53
Galectin-3	ng/mL	14	173	0.52

Table 4. Potential Blood Markers of Disease

Criteria:

Good correlation of serum to tumor
High in tumor compared to adjacent tissue

		Concentration			Correlation (r^2)
Analytes	Units	Adjacent	Serum	Tumor	Serum to Tumor
Osteopontin	ng/mL	0.0	26	1.6	1.0
IP-10	pg/mL	60	647	599	0.71
YKL-40	ng/mL	0.40	42	2.6	0.60
uPA	pg/mL	278	905	1660	0.53

Summary

- In CRC and PC tumor lysates,102 of the 115 proteins showed levels above the LLOQ. Figure 1 and Table 1 show 4 markers that were significantly higher in PC and 10 that were greater in CRC.
- For most of the analytes, duplicate sections of the tumor were similar, although some analytes did show differences. In 4 of the CRC analytes, Tumor 4 showed differences for CEA and tumor 2 for uPA. (Figure 2)
- Table 2 shows 30 analytes that were different in CRC tumor compared to its adjacent tissue. 10 of the analytes were higher in adjacent tissue compared to CRC.
- Eighteen of the markers examined showed significant correlations of CRC tumor concentration to serum levels. (Table 3) This suggests that they would be good protein markers to follow changes in tumor levels.
- The ideal serum biomarker of tumor activity would show good correlation to tumor levels and not to control tissue, as shown in Table 4.
- This platform provides a good method for studying changes in tumor levels as many proteins can be assessed with a very small sample.

Acknowledgements

This project has been funded with funds from the Cancer Prevention and Research Institute of Texas (CPRIT).